A-038

SSR240600 AND SAREDUTANT, NK1 AND NK2 RECEPTOR ANTAGONISTS RESPECTIVELY, INCREASE SOCIAL INTERACTION IN RATS IN A MODEL SENSITIVE TO ANXIOLYTIC DRUGS

Denis Boulay, Ronan Depoortere, Olivier Bergis and Guy Griebel
Sanofi-Aventis, CNS Research Department, Bagneux, France.

Introduction

- Tachykinins are a class of neuropeptides that have been shown to be involved in the modulation of emotional processes. This is notably illustrated by findings showing that tachykinin receptor ligands, such as neurokinin NK1 or NK2 antagonists are able to reduce stress-related behaviors in animals. For example, the nonpeptide NK1 receptor antagonist, SSR240600 [(R)-2-(1-{2-[4-{2-[3,5-bis(trifluoromethyl)phenyl]acetyl}-2-(3,4 dichlorophenyl)-2-morpholinyl]ethyl}-4-piperidinyl)-2 methylpropanamide], was found to inhibit distress vocalizations produced by maternal separation in guinea pig pups and to counteract the increase in body temperature induced by isolation stress. Studies with the NK2 receptor antagonist, SR48968 (saredutant), (S)-N-methyl-N[4-(4-acetylamino-4 phenylpiperidino)-2-(3,4-dichlorophenyl)-butyl] benzamide, revealed that the drug displayed anxiolytic-like effects in a variety of animal models, including the light/dark exploration and elevated plus-maze procedures, and the mouse defense test battery. The social interaction procedure is used to detect potential anxiolytic-like properties of psychoactive agents by measuring the level of social behaviors between pairs of rats under aversive /anxiogenic conditions (i.e. unfamiliar environment and high illumination) (File and Hyde, 1978). Under these latter conditions, in which the level of spontaneous interaction between two rats are low, anxiolytics increase social interaction. In the present study we investigated further the anxiolytic-like properties of SSR240600 and saredutant. Their effects were compared to those of the prototypical anxiolytics diazepam and buspirone, and the antidepressant, fluoxetine.

Methods

Animals

- Experiments were performed using rats, in accordance with the "Guide and Care and Use of Laboratory Animals" (National Institute of Health) and were approved by the in-house Animal Ethics Committee.

The social interaction test

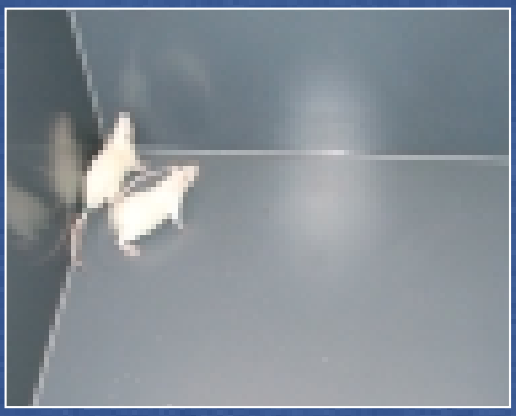

- The social interaction test arena (Photo 1) consisted of a grey Plexiglas box (75 x 75 x 42 cm high) with four white lights located above and on the side of the arena (delivering 235 lux at the level of the floor). A camera fixed above the arena was connected to a computer using the Ethovision, software (Noldus, Ethovision-Pro version 2.3), which allowed the tracking of each animal of the dyad. To that end, the system recorded the xy coordinates of the isobaric center of each rat. The system considered that a social interaction episode was taking place between the two rats whenever the distance between the two isobaric centers was less than 14 cm (user-defined distance, corresponding to the length between the head and the basis of the tail for rats weighing 180-200 g. Pairs of unfamiliar rats (from two different home cages) were treated intraperitonealy or subcutaneously (buspirone) (same treatment for both rats of the dyad: vehicle or one dose of the challenge compound) and isolated individually for 30 min. The pair was then placed into the arena during 10 min for social interaction recording. Data are expressed as the mean time spent in social interaction measured for each dyad of rats, and were analyzed by one-way ANOVA's with drug treatment as the between-groups factor, and followed, when appropriate, by post-hoc Dunnett's tests.

Discussion

- The present findings demonstrate that the NK1 and NK2 receptor antagonists, SSR240600 and saredutant, respectively, display anxiolytic-like effects in the social interaction test in rats. The magnitude of the effects of saredudant was comparable to those of diazepam and buspirone, two clinically effective anxiolytic agents, which were tested in parallel. The lack of effect of fluoxetine may at first glance be surprising since the drug is used clinically as an anxiolytic agent. However, very few animal studies were able to show anxiolytic-like effects of fluoxetine after an acute dosing, suggesting that chronic treatment may be required to produce anxiolysis. Taken together, these findings :
 - corroborate and extend previous results on the involvement of the central tachykinergic system in the modulation of emotional processes,
 - confirm that both the NK1 and NK2 receptor subtypes are involved in these effects,
 - suggest that NK1 and NK2 receptor antagonists may be useful for the treatment of pathological anxiety states.

Results

Figure 1 EFFECTS OF THE NK1 RECEPTOR ANTAGONIST SSR240600 IN THE SOCIAL INTERACTION TEST IN RATS

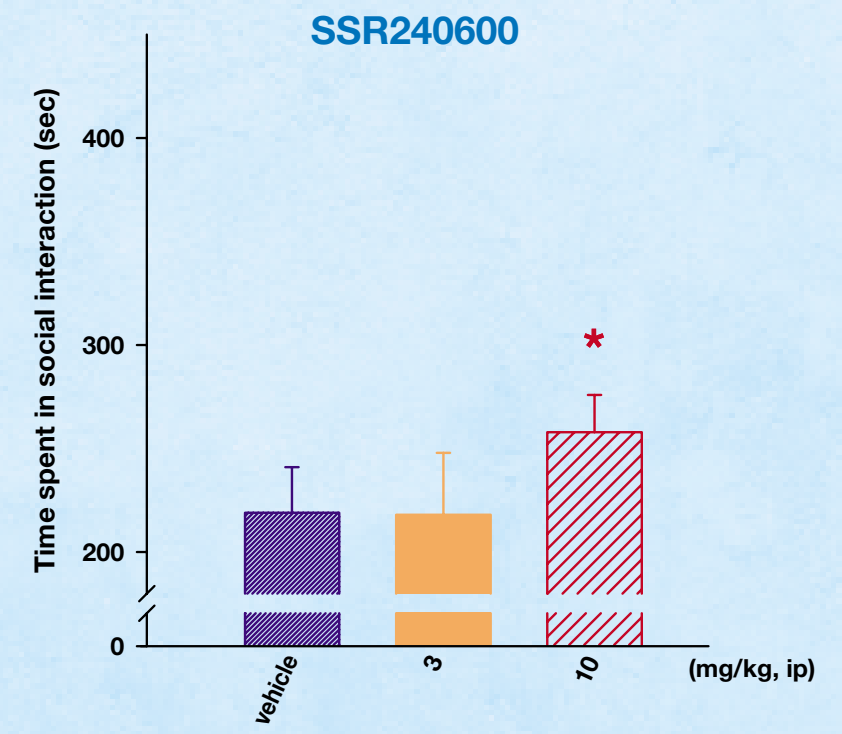
SSR240600
Time spent in social interaction (sec)
400
300
200
0
*
vehicle
3
10
(mg/kg, ip)

SSR240600 increased significantly the time spent in social interaction at 10 mg/kg.

Figure 2 EFFECTS OF THE NK2 RECEPTOR ANTAGONIST SAREDUDANT IN THE SOCIAL INTERACTION TEST IN RATS

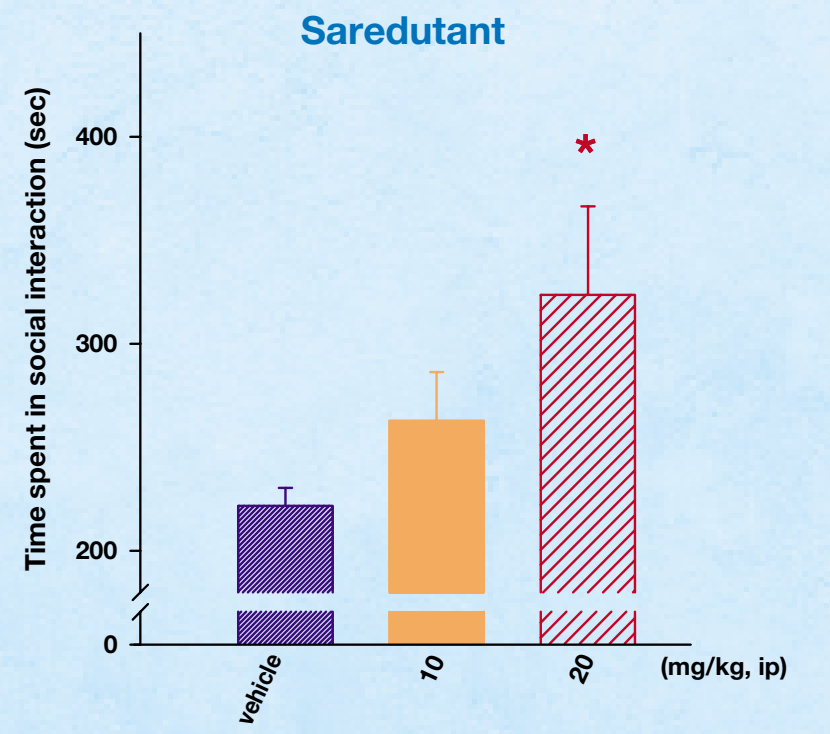
Saredutant
Time spent in social interaction (sec)
400
300
200
0
*
vehicle
10
20
(mg/kg, ip)

Saredudant increased significantly the time spent in social interaction at 20 mg/kg.

Figure 3 EFFECTS OF THE BENZODIAZEPINE ANXIOLYTIC DIAZEPAM IN THE SOCIAL INTERACTION TEST IN RATS

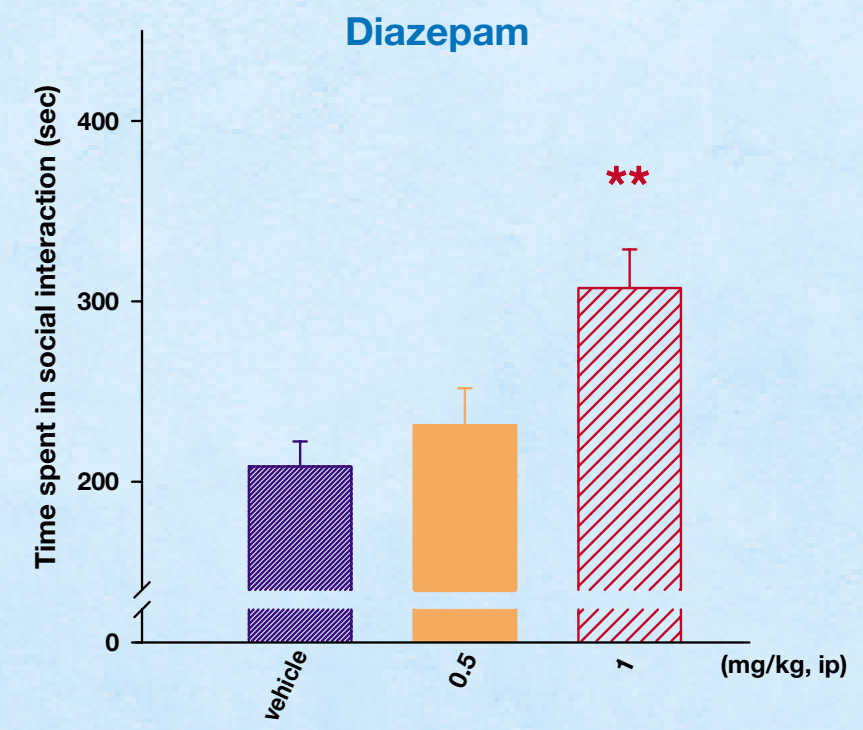
Diazepam
Time spent in social interaction (sec)
400
300
200
0
**
vehicle
0.5
1
(mg/kg, ip)

Diazepam increased significantly social interaction at 1 mg/kg.

Figure 4 EFFECTS OF THE 5-HT1A RECEPTOR AGONIST BUSPIRONE IN THE SOCIAL INTERACTION TEST IN RATS

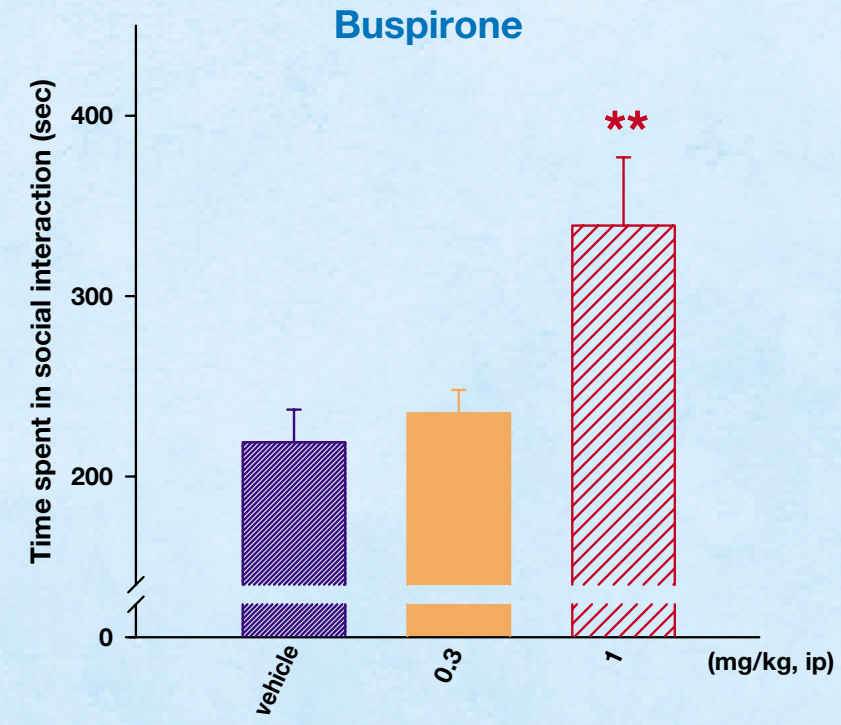
Buspirone
Time spent in social interaction (sec)
400
300
200
0
**
vehicle
0.3
1
(mg/kg, ip)

Buspirone increased significantly social interaction time at 1 mg/kg.

Figure 5 EFFECTS OF THE ANTIDEPRESSANT FLUOXETINE IN THE SOCIAL INTERACTION TEST IN RATS

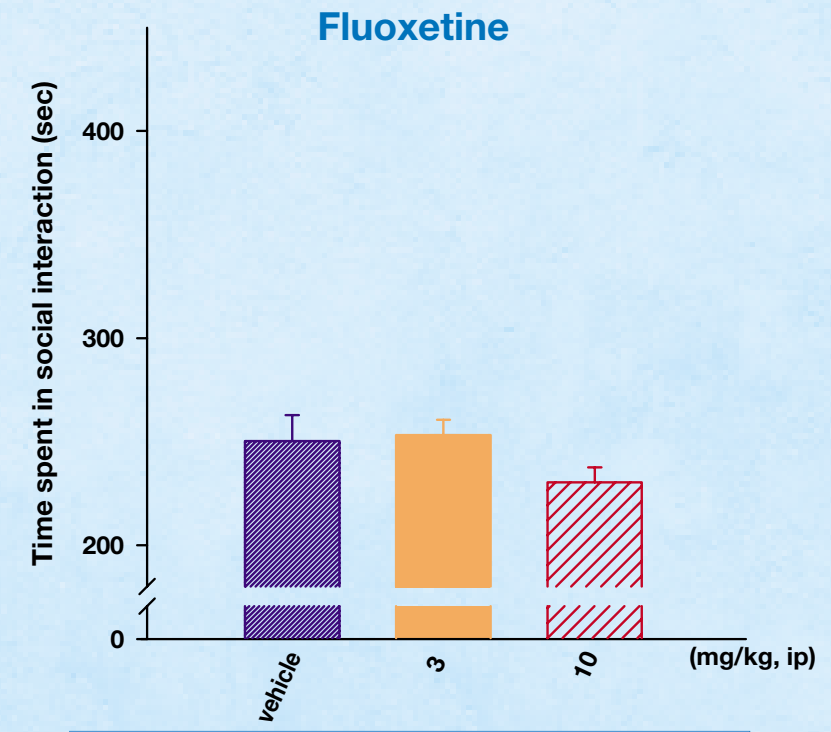
Fluoxetine
Time spent in social interaction (sec)
400
300
200
0
vehicle
3
10
(mg/kg, ip)

Fluoxetine did not modify significantly the level of social interaction.

Acknowledgements

We are grateful to technical assistance of Martine Lacave.

11th Biennial European Behavioural Pharmacology Society Meeting • September 9-12, 2005, Barcelona, Spain

EQUINOXE - 0805