Effects of increasing adult hippocampal neurogenesis in mice during exposure to chronic stress

636.02

L. CULIG[1,2], S. LEGENDRE[1,2], F. MINIER[1,2], G. GRIEBEL[3], A. SAHAY[4,5], R. HEN[6], C. BELZUNG[1,2]

[1]Univ. François Rabelais, Tours, France; [2]Inserm U930, Tours, France; [3]Evaluation & Expertise, Sanofi, Chilly-Mazarin, France; [4]Ctr. for Regenerative Med., Boston, MA; [5]Harvard Stem Cell Inst., Boston, MA; [6]Div. of Integrative Neuroscience, Departments of Neurosci. and Psychiatry, Columbia Univ., New York, NY

AIM OF THE STUDY

- Investigate depressive-like behavior
- Mechanism: Long-term neuronal activity[1] in the NAc and other areas?

After increasing adult hippocampal neurogenesis

WHAT WE WANTED TO DO

Increase neurogenesis during unpredictable chronic mild stress and behaviorally test these mice (with a focus on anhedonic features) when neurogenesis is at its peak

WHY?

Increasing neurogenesis is sufficient to reduce some of depressive-like behaviors[2] in mice, but the mechanism has not been elucidated.

HOW?

Using a transgenic mouse line (*iBax*), in which neural stem cells apoptosis was diminished, enhancing survival and functional integration of new born neurons in the adult brain.

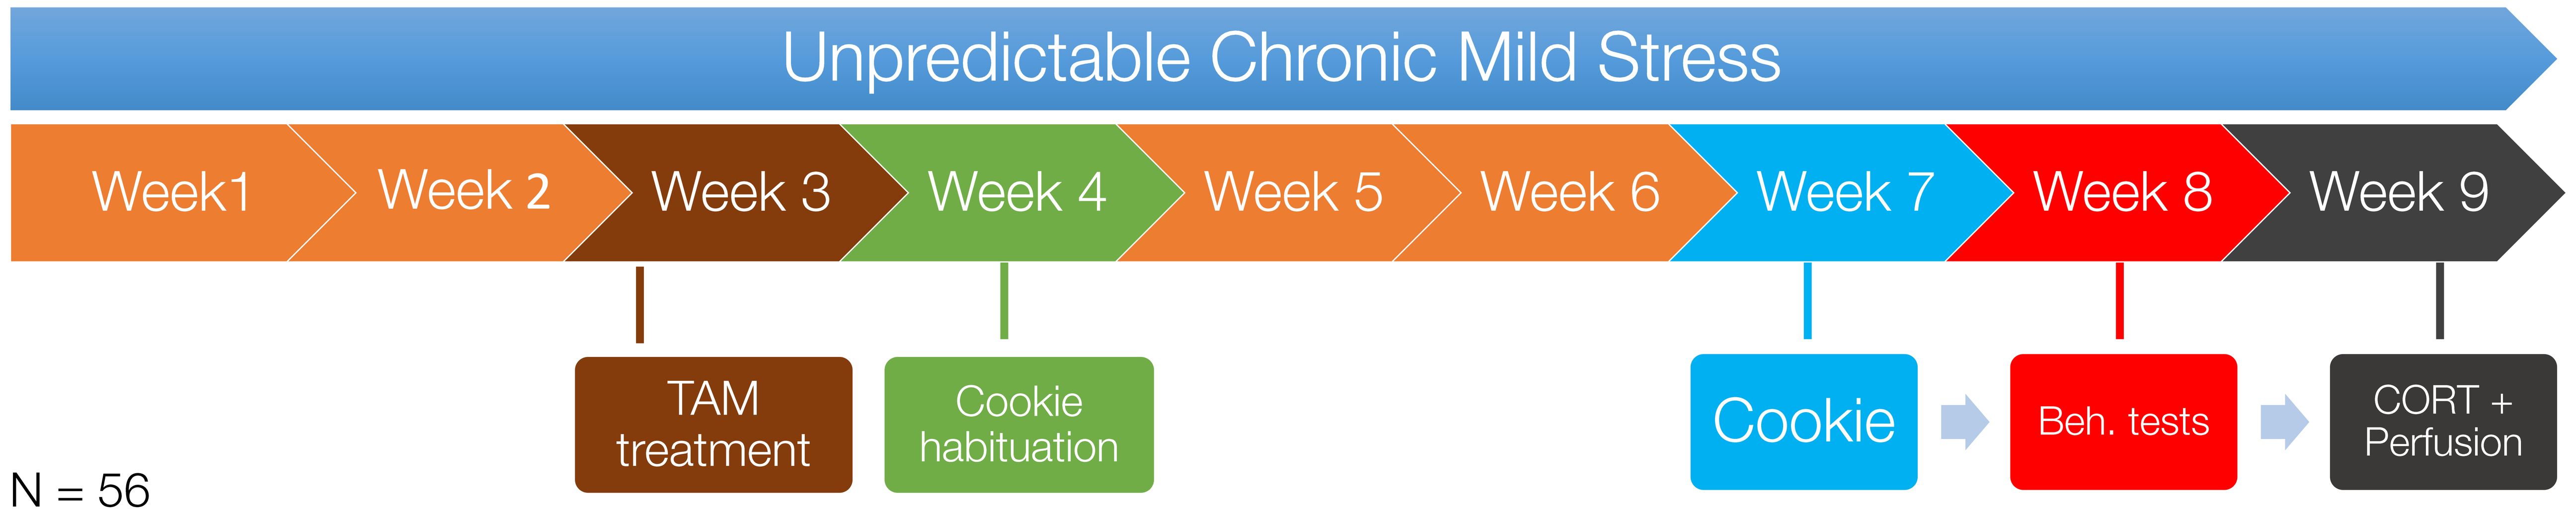
Unpredictable Chronic Mild Stress
Week1
Week 2
Week 3
Week 4
Week 5
Week 6
Week 7
Week 8
Week 9
TAM treatment
Cookie habituation
Cookie
Beh. tests
CORT + Perfusion
N = 56

INTRODUCTION

Local changes in the hippocampal network (addition of new neurons in the dentate gyrus) might change the activity of neural circuitry in the areas to which the hippocampus projects. Among those structures are the mPFC (medial prefrontal cortex), amygdala and nucleus accumbens (NAc), which has a crucial role in reward and motivation. There are two notable effects of chronic stress on neurogenesis (dcx ↓) and ΔFosB (↑).

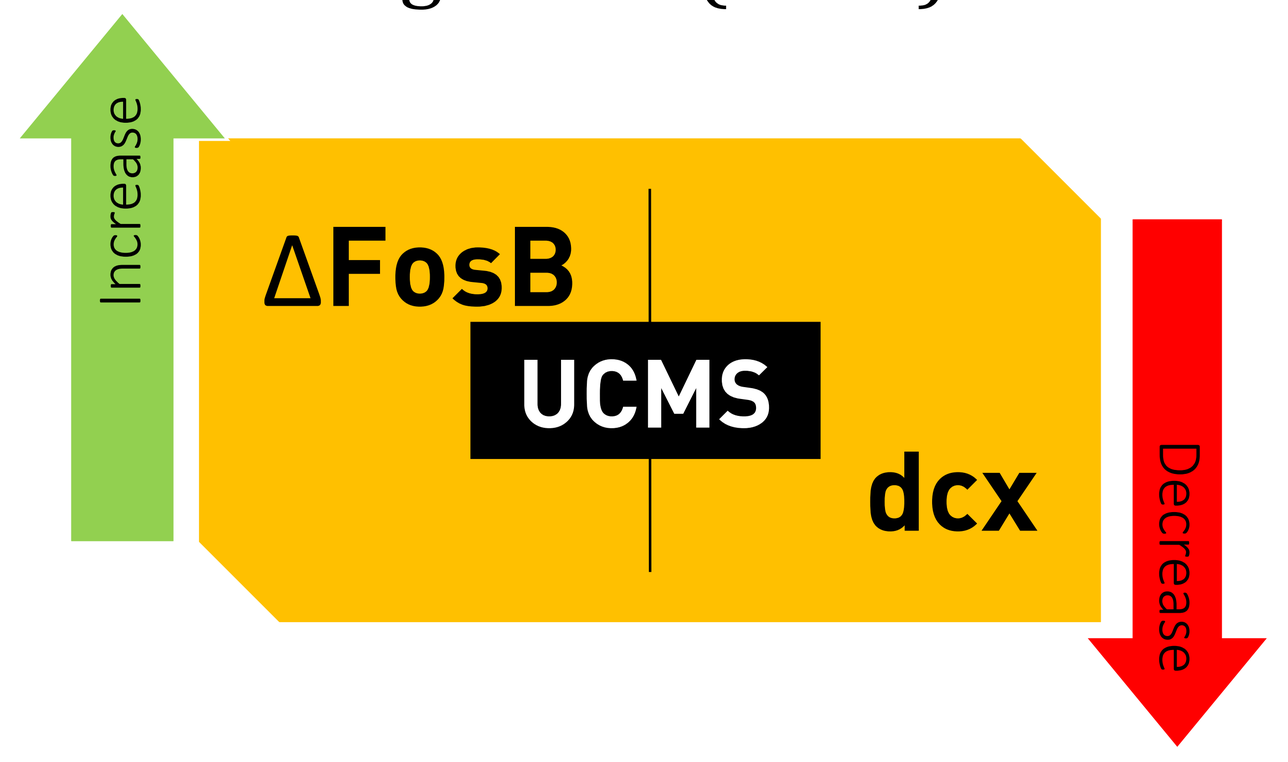
Increase
ΔFosB
UCMS
dcx
Decrease

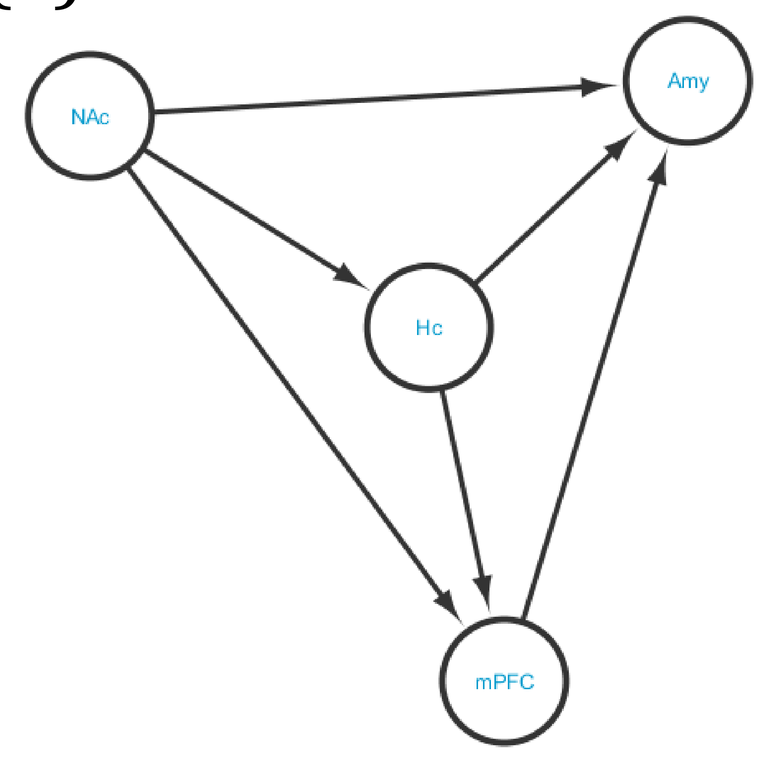
NAc
Amy
Hc
mPFC

Accumulation of ΔFosB, an unusually stable transcription factor which accumulates over time after repeated stress exposure, has been observed in many animal models of depression, including unpredictable chronic mild stress[3] (UCMS), and it could be the basis of certain behavioral consequences of exposure to chronic stress. This ΔFosB induction could have a protective role against stress, but no studies so far have explored how a specific increase in neurogenesis might regulate the induction.

The main aim of this study is to test this hypothesis by using *iBax* mice, in which the pro-apoptotic gene *Bax* was selectively ablated in neural stem cells, inducibly enhancing survival and functional integration of new born neurons in the adult brain. The animals were exposed to UCMS, a naturalistic rodent model of depression, and then tested on behavioral, physiological and histological levels to assess depressive-live and anxiety-like behavior, the HPA axis activity and neuronal activation in the NAc.

MATERIALS AND METHODS

We subjected *iBax* mice aged from 11 to 14 weeks to 9 weeks of UCMS, consisting of randomized social and environmental stressors, in order to induce a depressive-like state[4].

In week 3 of the UCMS protocol, we treated the animals with tamoxifen (TAM) for five days (55 mg/kg/day, ip, in corn oil) in order to induce recombination of *Bax* in neural stem cells.

In week 7 animals went through 3 trials of the cookie test and in week 8, animals went through a batch of behavioral tests to assess traits related to depression and anxiety.

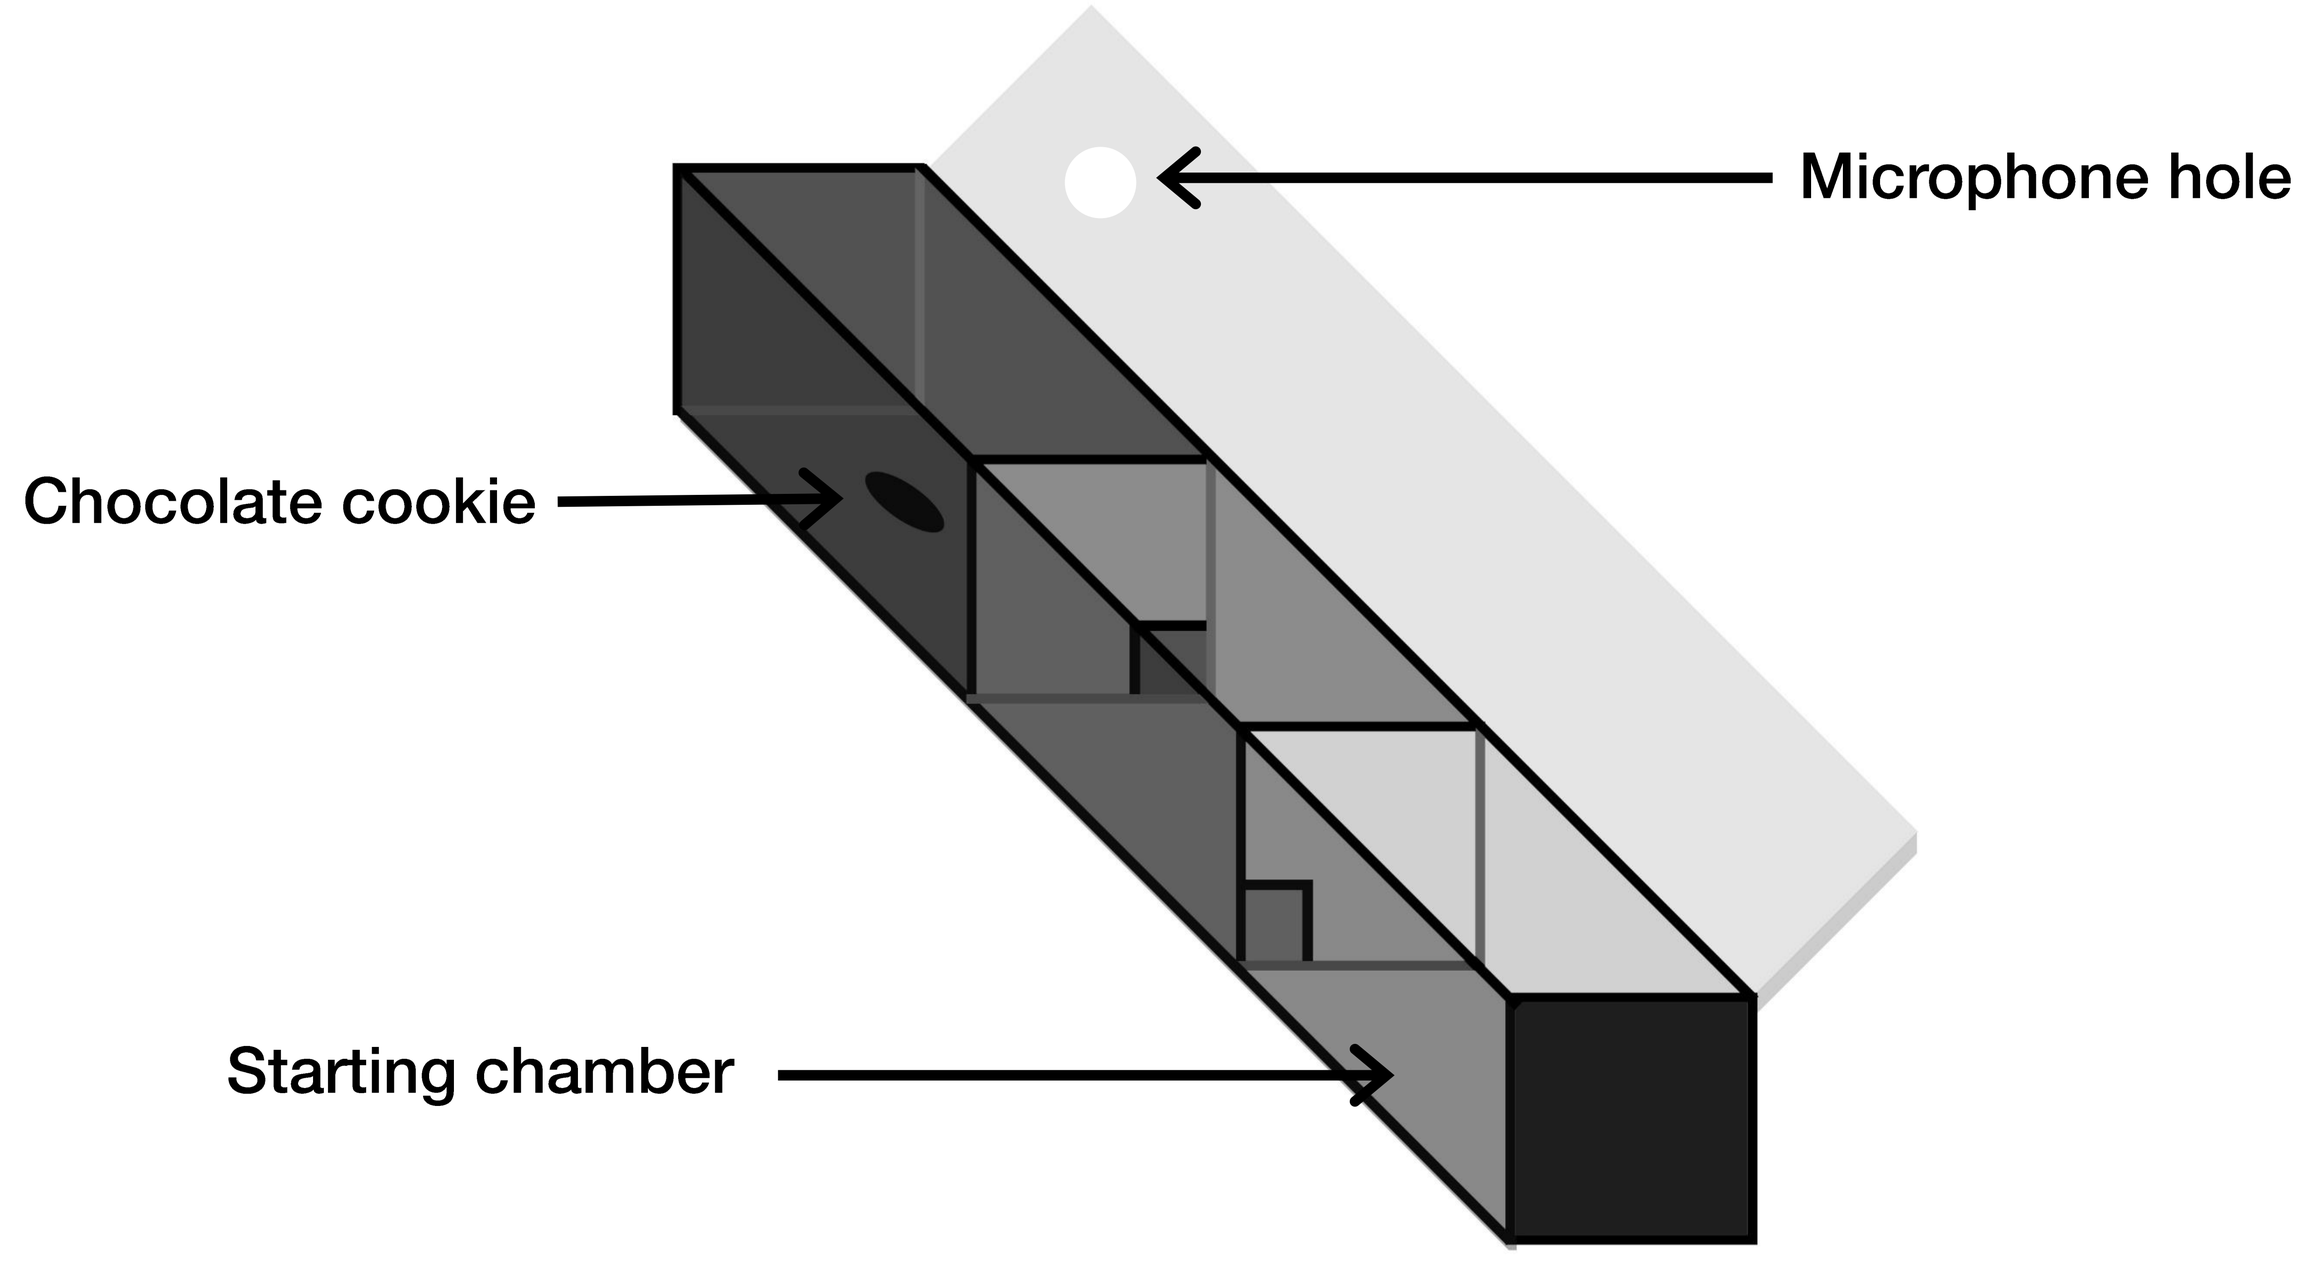
Microphone hole
Chocolate cookie
Starting chamber

In week 9, the animals had their blood collected to assess basal plasma corticosterone levels and the animals were either (1) transcardially perfused and their brains collected for ΔFosB immunohistochemistry or (2) killed by CO_2 asphyxiation and had NAc, mPFC and Amy dissected and frozen on dry ice for subsequent western blotting.

RESULTS

DCX

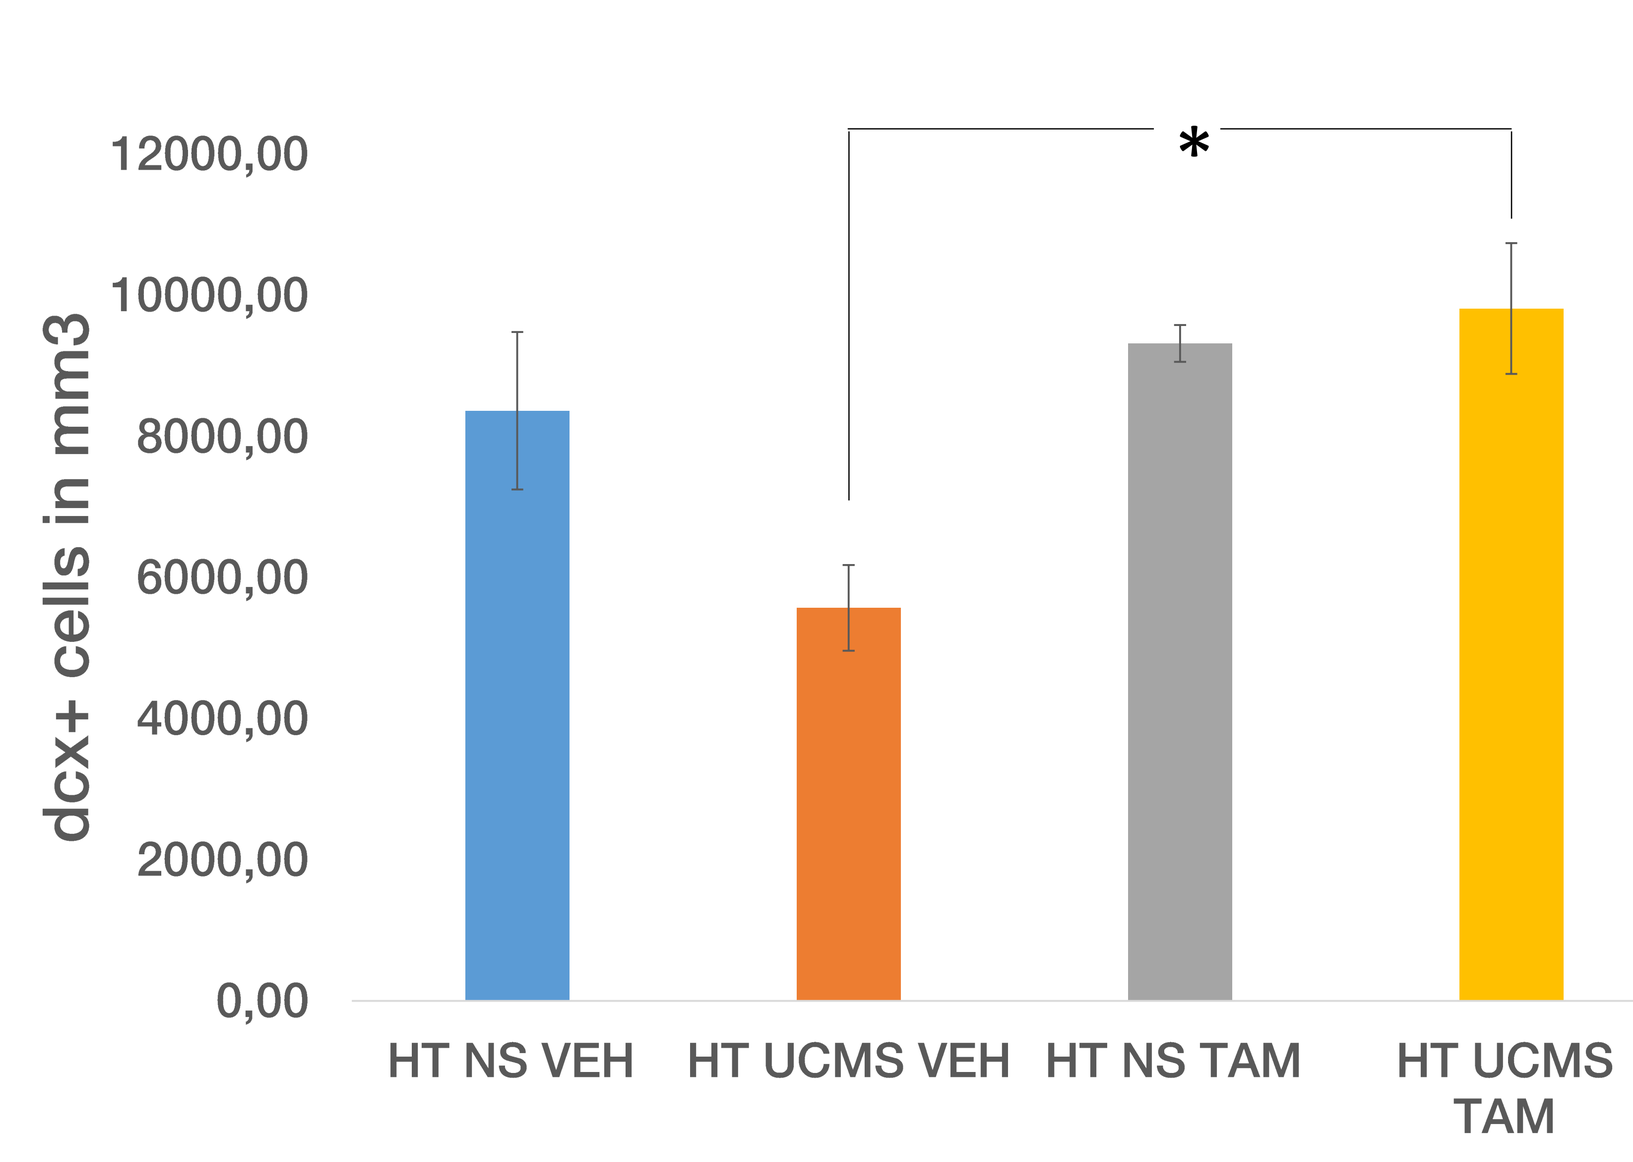
dcx+ cells in mm3
12000,00
10000,00
8000,00
6000,00
4000,00
2000,00
0,00
*
HT NS VEH
HT UCMS VEH
HT NS TAM
HT UCMS TAM

COOKIE TEST 3

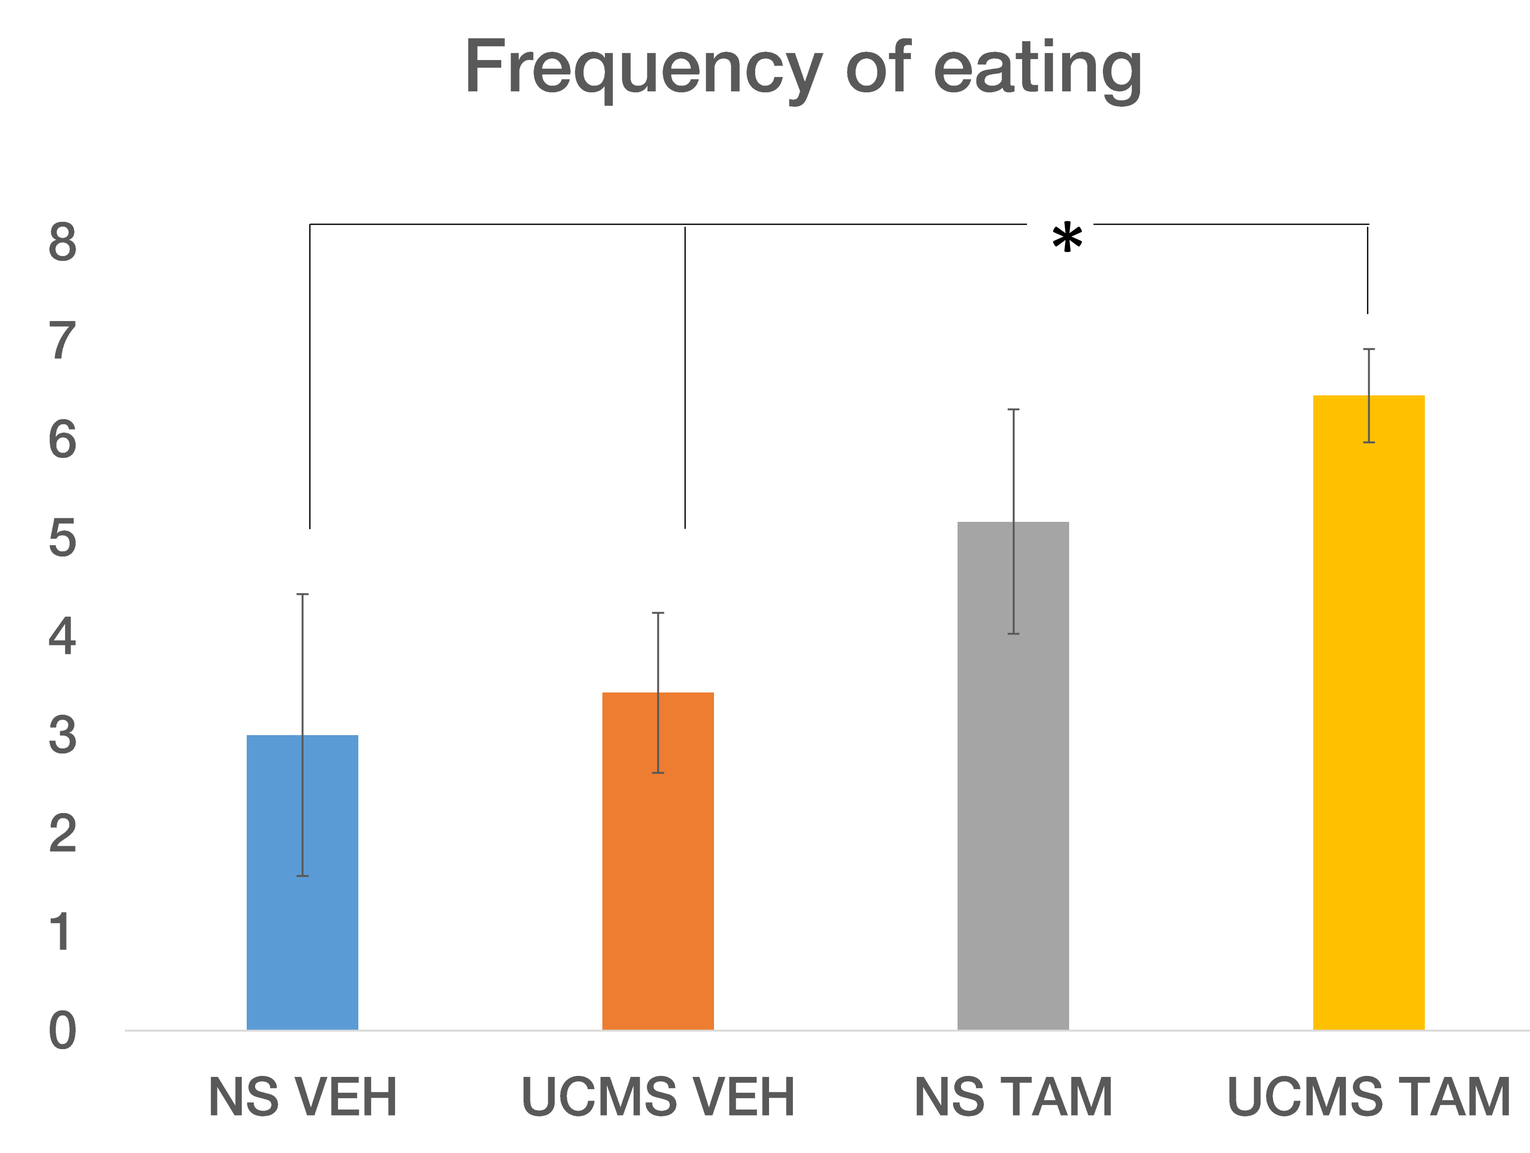
Frequency of eating
8
7
6
5
4
3
2
1
0
*
NS VEH
UCMS VEH
NS TAM
UCMS TAM

ΔFosB EXPRESSION IN THE NAc

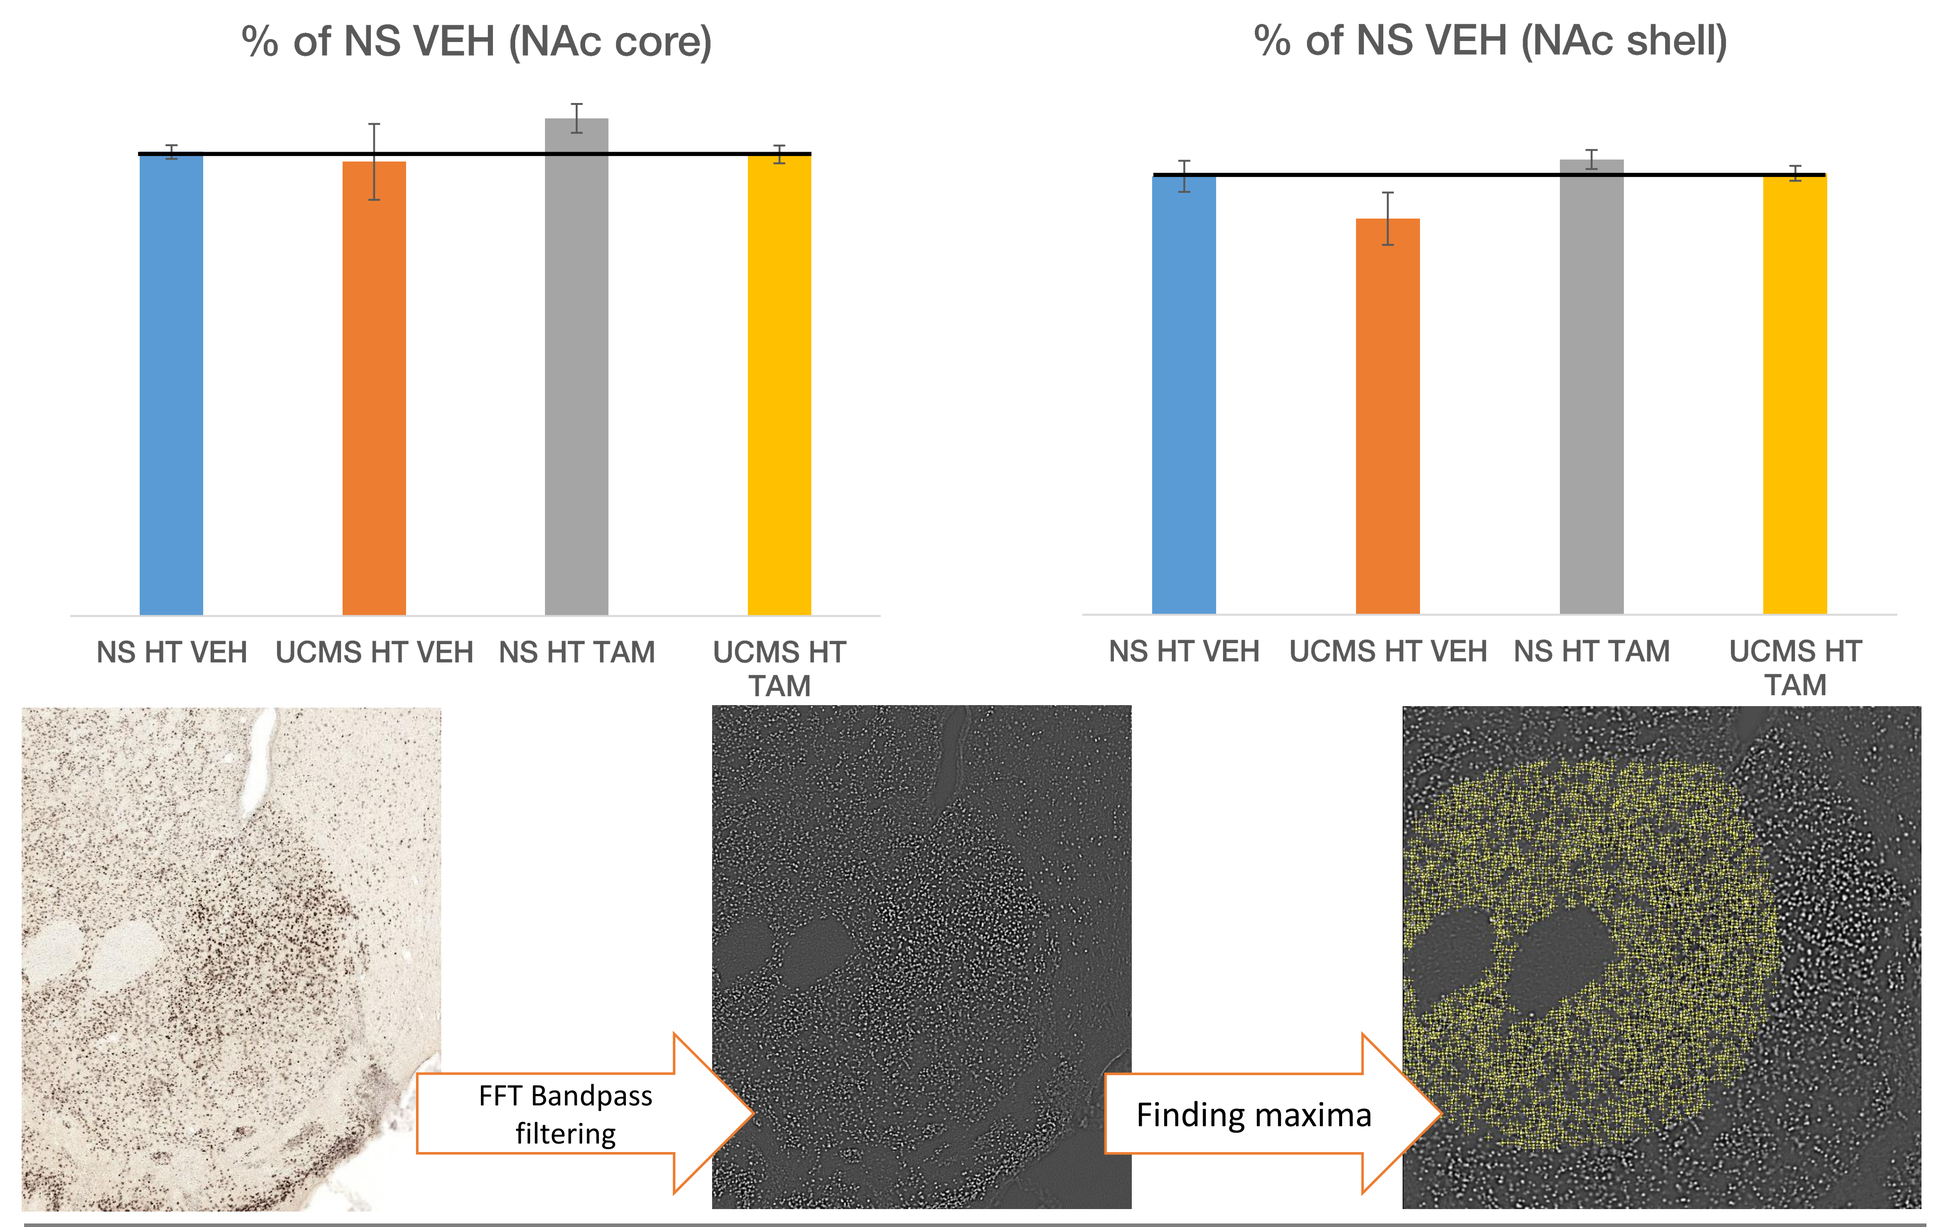
% of NS VEH (NAc core)
% of NS VEH (NAc shell)
NS HT VEH
UCMS HT VEH
NS HT TAM
UCMS HT TAM
FFT Bandpass filtering
Finding maxima

COAT STATE

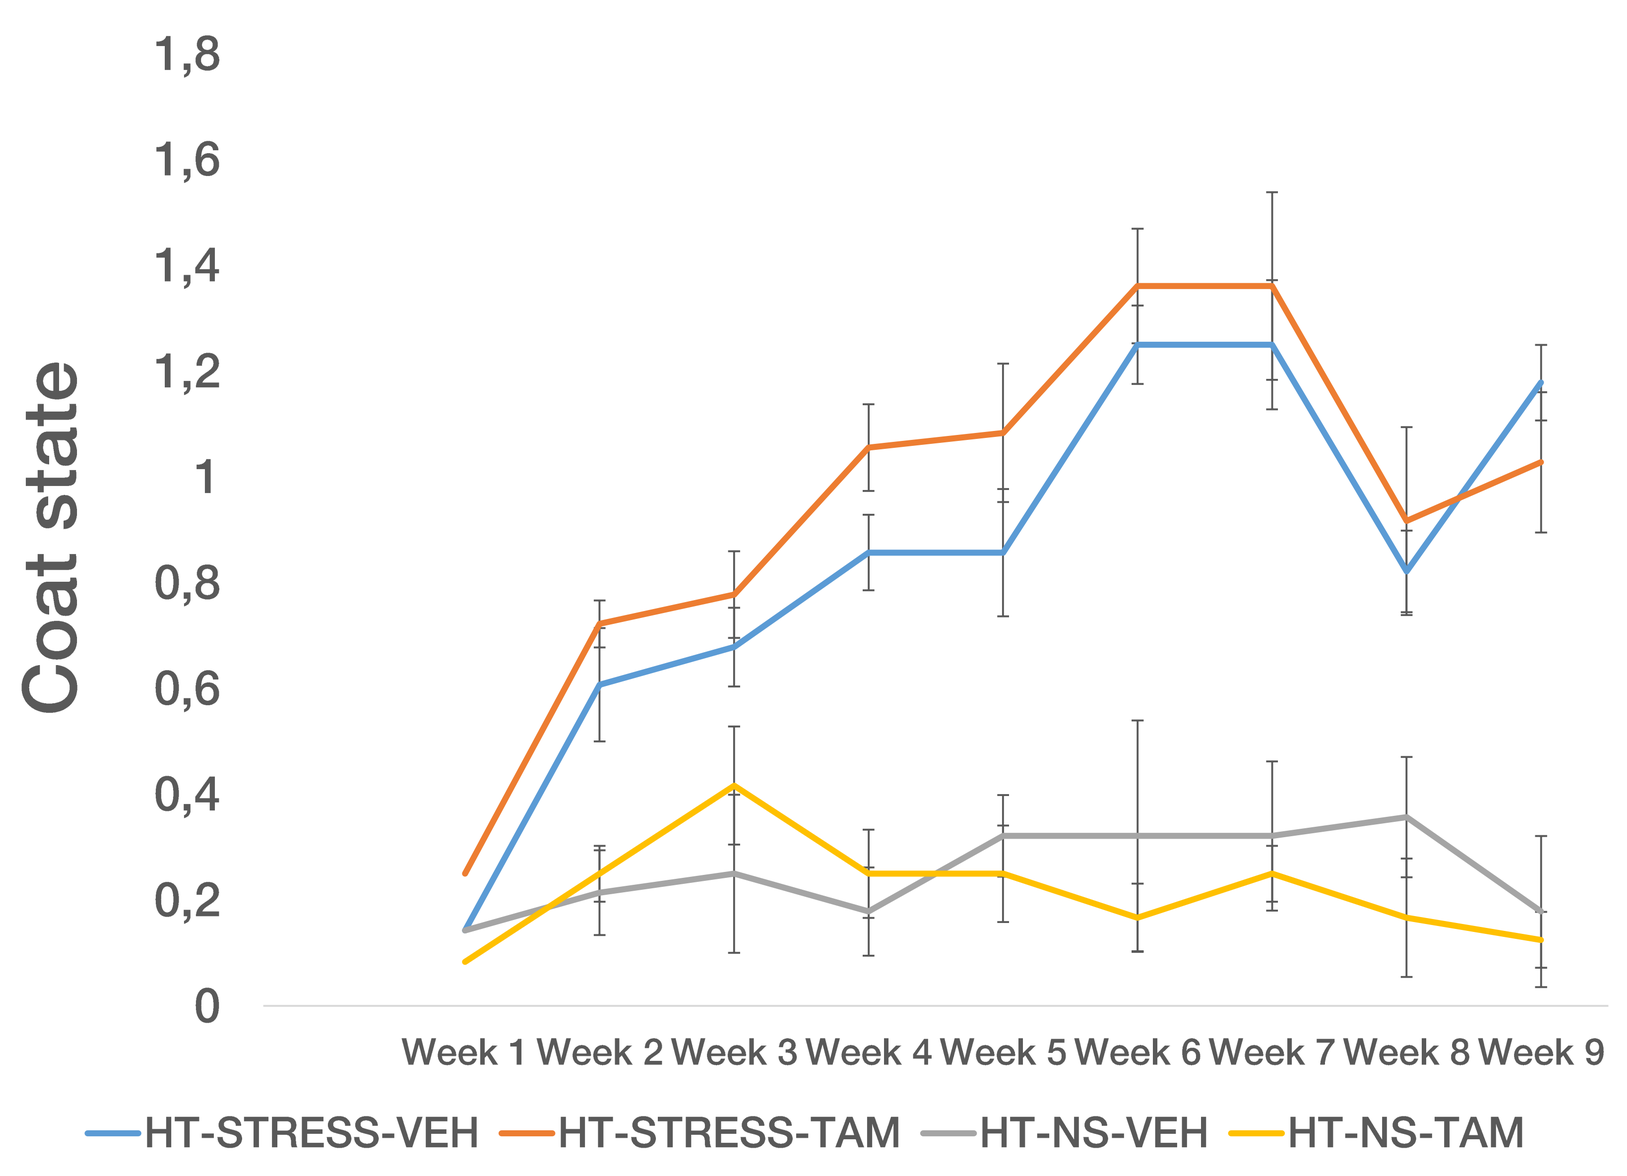
Coat state
1,8
1,6
1,4
1,2
1
0,8
0,6
0,4
0,2
0
Week 1 Week 2 Week 3 Week 4 Week 5 Week 6 Week 7 Week 8 Week 9
HT-STRESS-VEH
HT-STRESS-TAM
HT-NS-VEH
HT-NS-TAM

LIGHT/DARK BOX

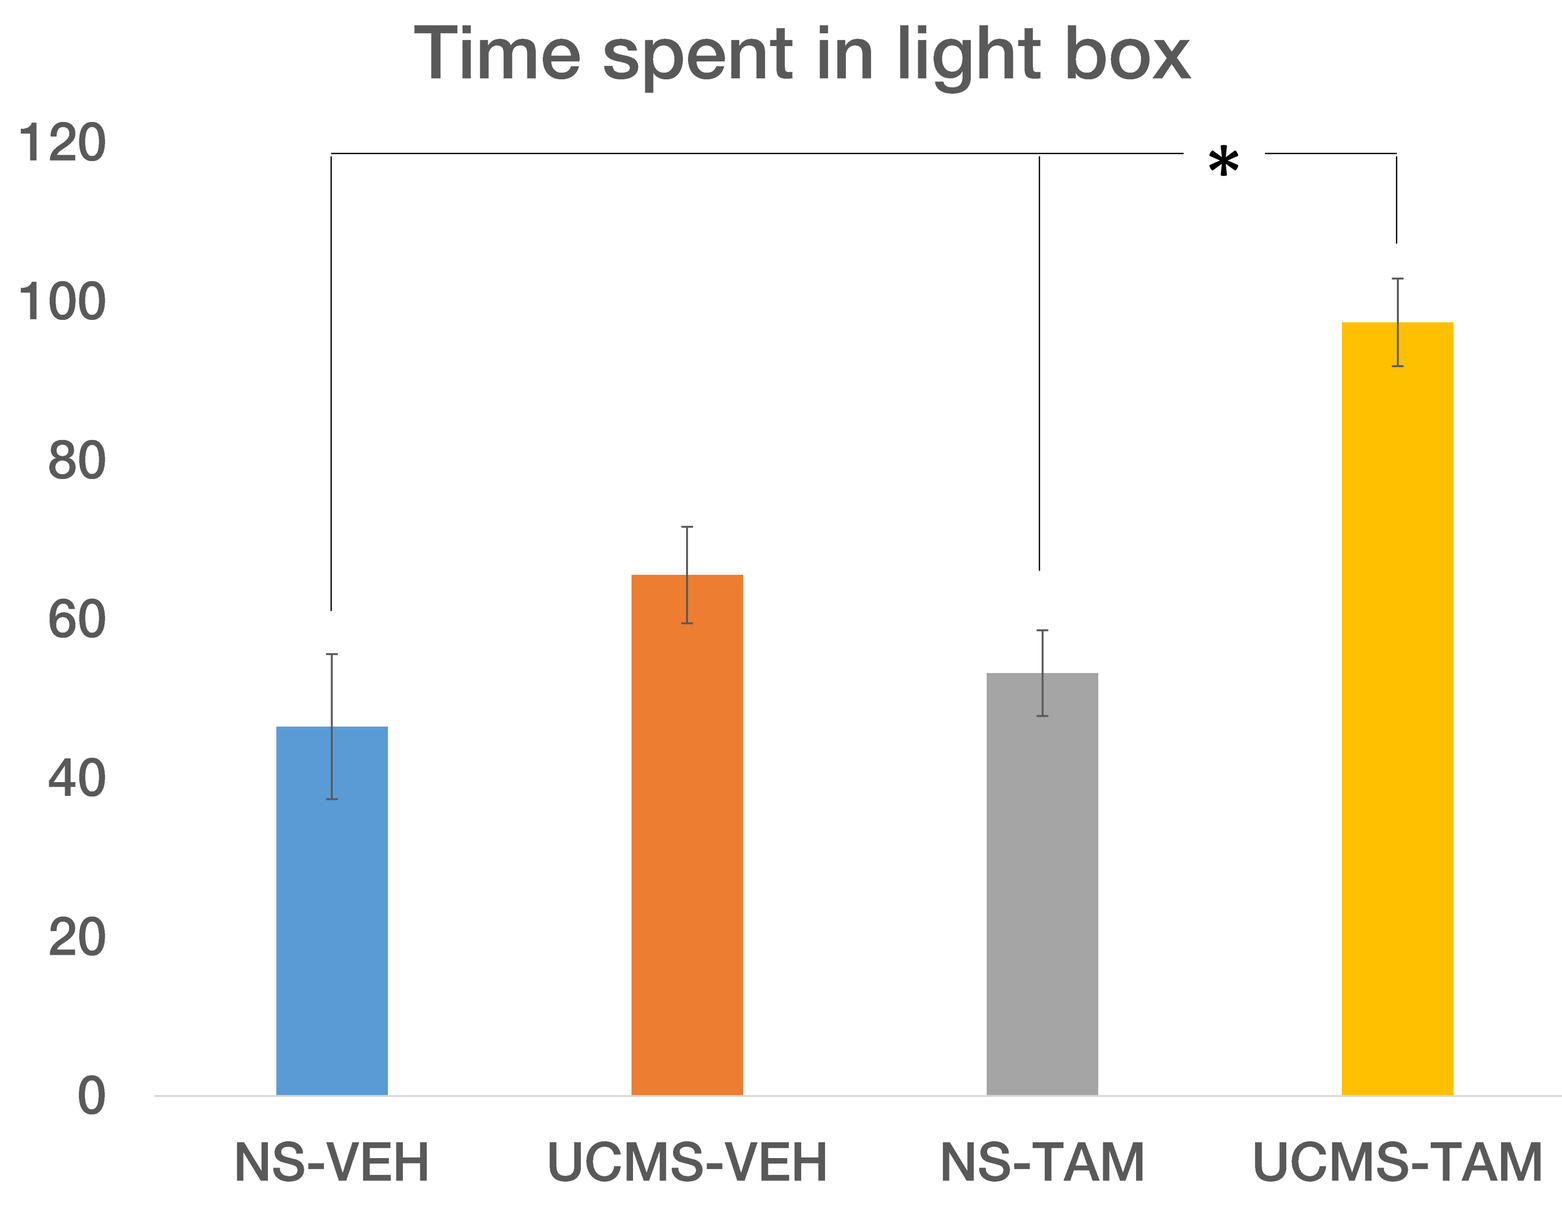
Time spent in light box
120
100
80
60
40
20
0
*
NS-VEH
UCMS-VEH
NS-TAM
UCMS-TAM

BASAL CORTICOSTERONE

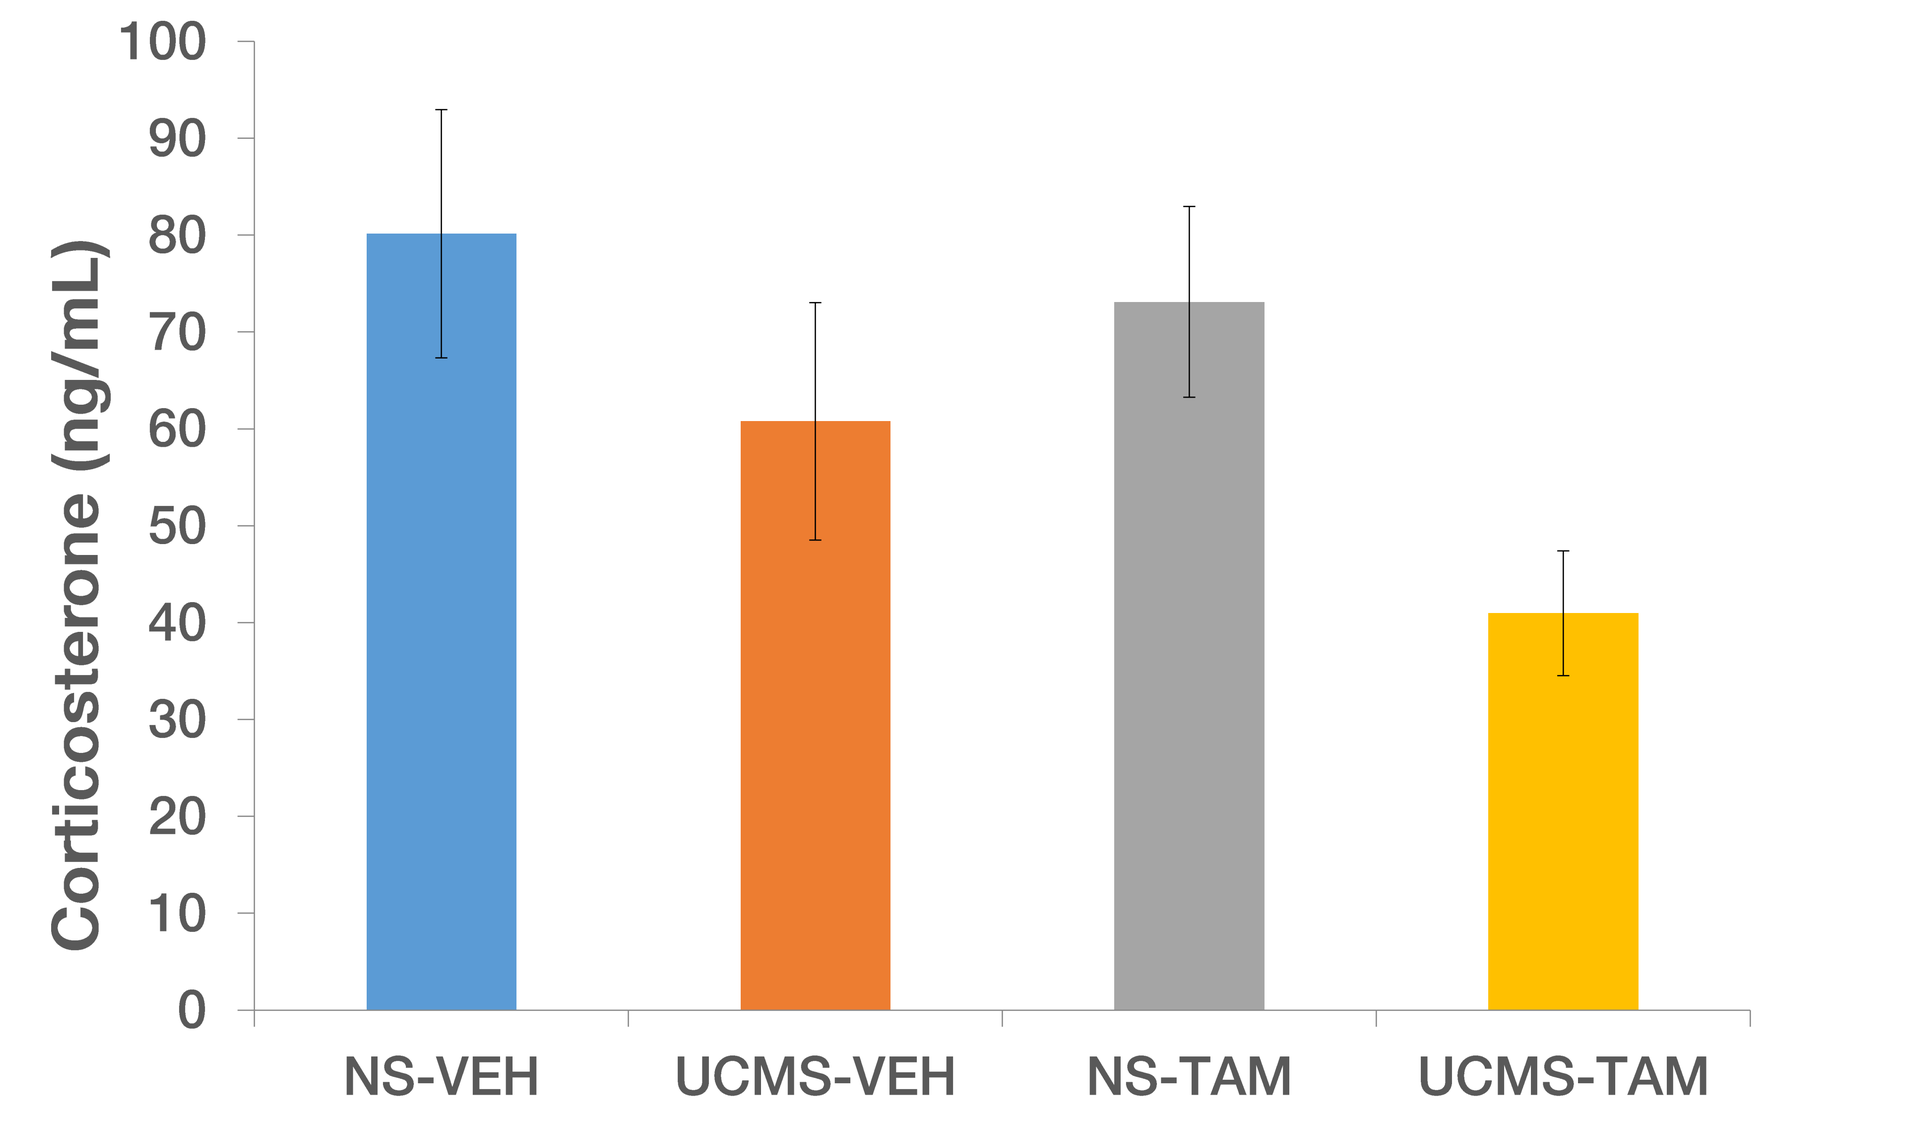
Corticosterone (ng/mL)
100
90
80
70
60
50
40
30
20
10
0
NS-VEH
UCMS-VEH
NS-TAM
UCMS-TAM

DISCUSSION AND CONCLUSIONS

Tamoxifen treatment induced an increase of neurogenesis in mice aged 13-16 weeks at the time of the injections. We observed an increase in the cookie consumption in the cookie test for the UCMS-TAM group, and such an increase may be related to hedonic features. We also observed a difference in the light/dark box test, where the UCMS-TAM group spent significantly more time in the light box, pointing to a less anxious phenotype. However, we did not detect a significant difference in coat state. No statistically significant difference was found between groups in locomotor activity, suggesting that the differences found in other tests are not due to different mobility.

No statistically significant difference was found between groups in basal corticosterone, suggesting that the HPA axis was functioning normally during baseline conditions. Also, we did not observe any significant difference in neuronal activity in the NAc (core and shell), which prompts us to perform a brain-wide ΔFosB expression analysis and functional network construction.

Overall, we found that increasing adult hippocampal neurogenesis provided a buffer against the effects of stress on the behavioral level in regards to anhedonia in anxiety-like behavior, and further analyses are underway to examine these findings in more detail.

REFERENCES

1. Vialou, V. *et al.* DeltaFosB in brain reward circuits mediates resilience to stress and antidepressant responses. *Nat. Neurosci.* **13,** 745–52 (2010)

2. Hill, A. S., Sahay, A. & Hen, R. Increasing Adult Hippocampal Neurogenesis is Sufficient to Reduce Anxiety and Depression-Like Behaviors. *Neuropsychopharmacology* 1–11 (2015).

3. Perrotti, L. I. *et al.* Induction of deltaFosB in reward-related brain structures after chronic stress. *J. Neurosci.* **24,** 10594–602 (2004).

4. Nollet, M., Le Guisquet, A.-M. & Belzung, C. Models of depression: unpredictable chronic mild stress in mice. *Curr. Protoc. Pharmacol.* **Chapter 5,** Unit 5.65 (2013)

UNIVERSITÉ FRANÇOIS - RABELAIS TOURS

Inserm

Neuroscience 2015

TOURS POITIERS Neuroimagerie Fonctionnelle SFR FED4226 Structure Fédérative de Recherche