CHARACTERIZATION OF THE CANNABINOID CB1 RECEPTOR ANTAGONIST SURINABANT (SR147778) IN MODELS OF COGNITION IN RODENTS

Philippe Pichat, Jean-Paul Terranova, Olivier Bergis, Guy Griebel, Bernard Scatton, Gérard Le Fur.
Sanofi-Aventis, Psychopharmacology Department, Bagneux, Montpellier, France

Introduction

- Several lines of evidence suggest that the central cannabinoid system is implicated in the modulation of cognitive processes, particularly learning and memory. Endocannabinoids and synthetic cannabinoid agonists produce memory dysfunctions in humans and in animals.

- The high concentration of CB1 receptors and the presence of endocannabinoids in the hippocampus and in cortical structures indicate that CB1 antagonists may modulate positively learning and memory processes. Disruptive effects of cannabinoids on short-term memory were shown in tests based either on conditioned or on spontaneous behavior. These effects were blocked by CB1 receptor antagonists, indicating that the cognitive-modulating action of cannabinoids is mediated by this receptor subtype. In line with this idea are findings with CB1 receptor knockout mice that showed improved memory performance in several tasks, including the object recognition and Morris water maze tests.

- In this context, the following experiments aimed at investigating the new selective CB1 receptor antagonist Surinabant (SR147778) in reliable rodent models predictive of therapeutic activity on cognitive symptoms related to schizophrenia or Alzheimer's disease.

Methods

OBJECT RECOGNITION TASK IN RATS

- The object recognition task is considered as a model of episodic memory in rodents.Twenty-four hours after an initial exposure to an open-field during which male Sprague-Dawley rats were allowed to habituate to the environmental context, animals were placed again in this apparatus in the presence of two identical objects (learning session) (see figure 1). Following a 1-or 24-hour forgetting delay, the retention (recall session) was evaluated by placing rats with the familiar object and a new one, and objects exploration duration was recorded. In the protocol using a pharmacologically-induced deficit and a short-term forgetting delay, Surinabant was administered intraperitoneally prior to the cognitive-impairing drug scopolamine, before the learning session. Using a long-term forgetting delay, treatment was given orally before each of the test sessions.

SOCIAL RECOGNITION TASK IN AGED MICE

- The social recognition procedure evaluates olfactory reference memory, which is independent of spatial context using aged (up to 12-month-old) mice confronted with a conspecific juvenile on two successive presentations with an intertrial interval of 30 min (see figure 4). The experiment was conducted using male C57BL/6 mice. Time spent in social interaction (nosing, sniffing, grooming the juvenile) was recorded. Surinabant (i.p.) was given following the first social learning presentation i.e. during the consolidation phase.

SPATIAL WORKING MEMORY TASK IN RATS

- The water maze protocol used permits the evaluation of spatial working memory acquisition by testing the latency of rats to find an invisible platform located in a circular arena (see figure 6). Adult male Sprague-Dawley rats were submitted to four 120-second trials per day (each trial separated by a 30-second interval). Each day, the platform location was changed to the center of each quadrant of the arena. All rats received two administrations of Surinabant (p.o.) and/or scopolamine (i.p.) or vehicle, 60 min and 30 min before the first trial of each day, respectively.

Results

OBJECT RECOGNITION TASK : EFFECT OF SURINABANT ON EPISODIC MEMORY IN RATS

Figure 1 EXPERIMENTAL DESIGN

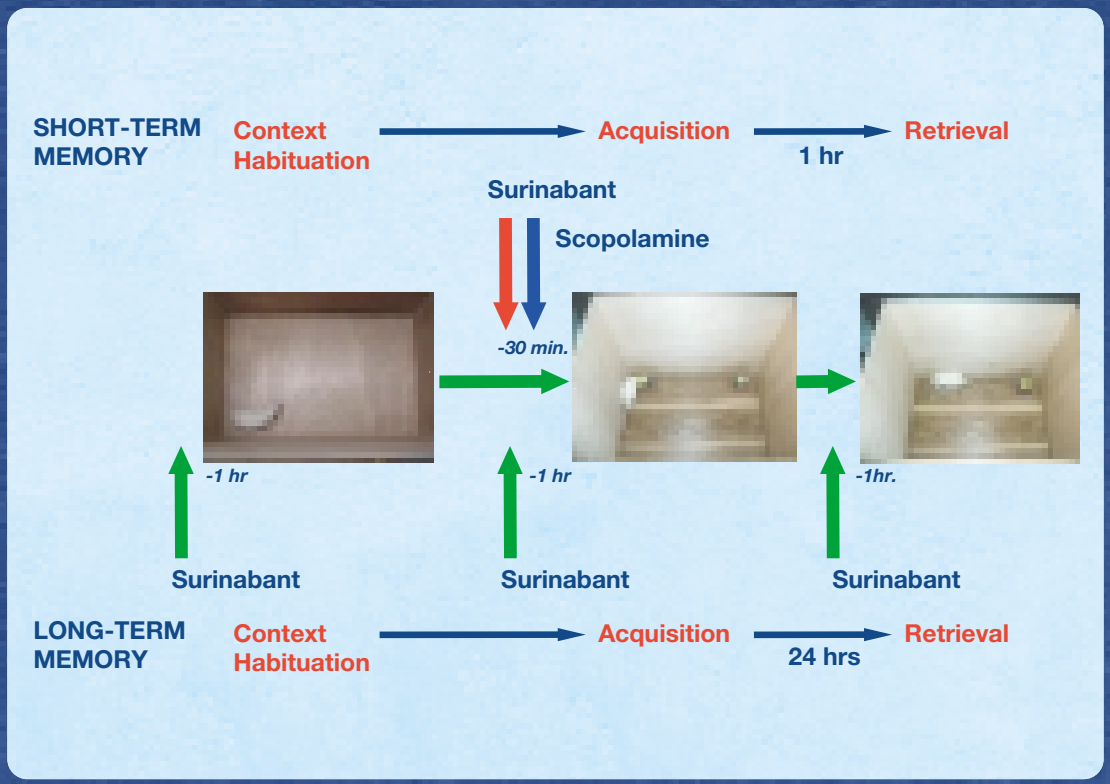
SHORT-TERM MEMORY
Context Habituation
Acquisition
1 hr
Retrieval
Surinabant
Scopolamine
-30 min.
-1 hr
-1 hr
-1hr.
Surinabant
Surinabant
Surinabant
LONG-TERM MEMORY
Context Habituation
Acquisition
24 hrs
Retrieval

Figure 2 REVERSAL BY SURINABANT OF SCOPOLAMINE-INDUCED EPISODIC MEMORY DURING RECALL

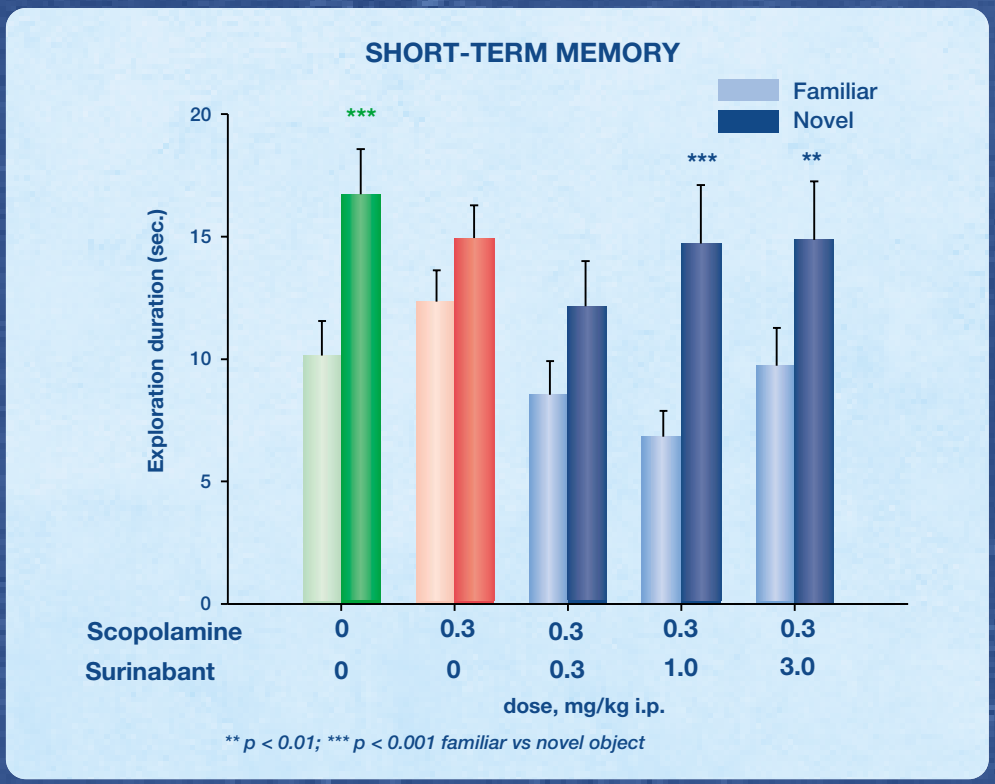
SHORT-TERM MEMORY
Familiar
Novel
Exploration duration (sec.)
Scopolamine 0 0.3 0.3 0.3 0.3
Surinabant 0 0 0.3 1.0 3.0
dose, mg/kg i.p.
** p < 0.01; *** p < 0.001 familiar vs novel object

Figure 3 ASSESSMENT OF PROMNESIC ACTIVITY OF SURINABANT

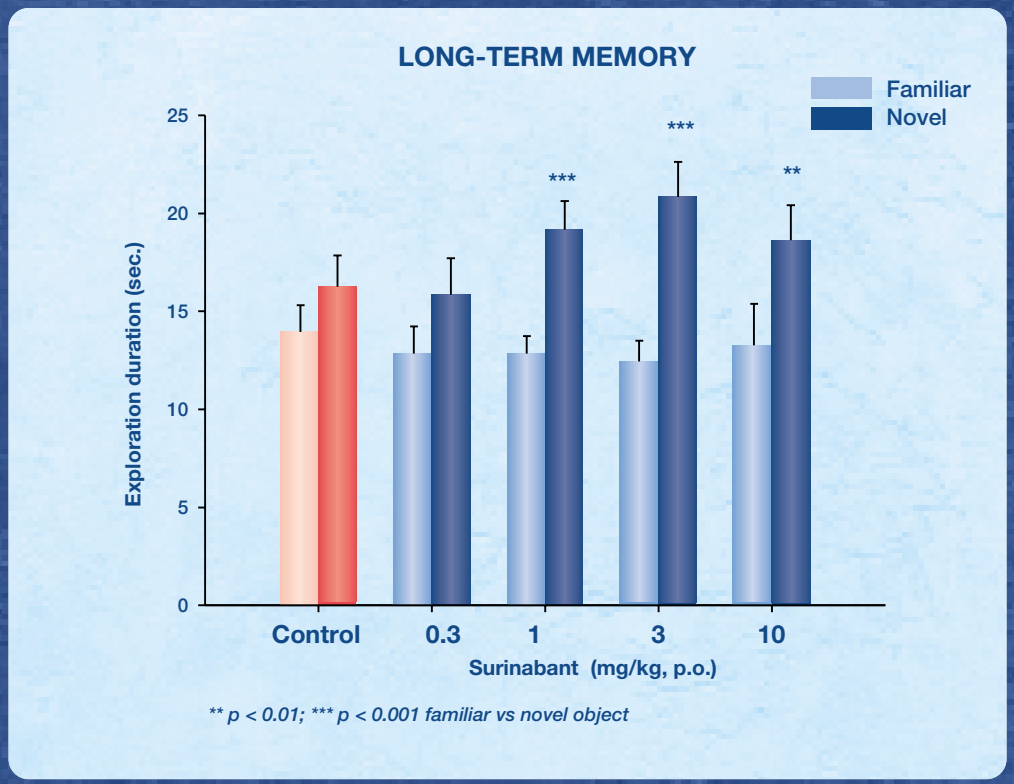
LONG-TERM MEMORY
Familiar
Novel
Exploration duration (sec.)
Control 0.3 1 3 10
Surinabant (mg/kg, p.o.)
** p < 0.01; *** p < 0.001 familiar vs novel object

Surinabant restores short-term episodic memory deficit induced by scopolamine from 1 mg/kg, i.p.
In addition, it also markedly improves long-term episodic memory from 1 mg/kg p.o. in the object recognition tasks

SOCIAL RECOGNITION TASK : EFFECT OF SURINABANT ON SOCIAL MEMORY DEFICIT IN AGED MICE

Figure 4 EXPERIMENTAL DESIGN

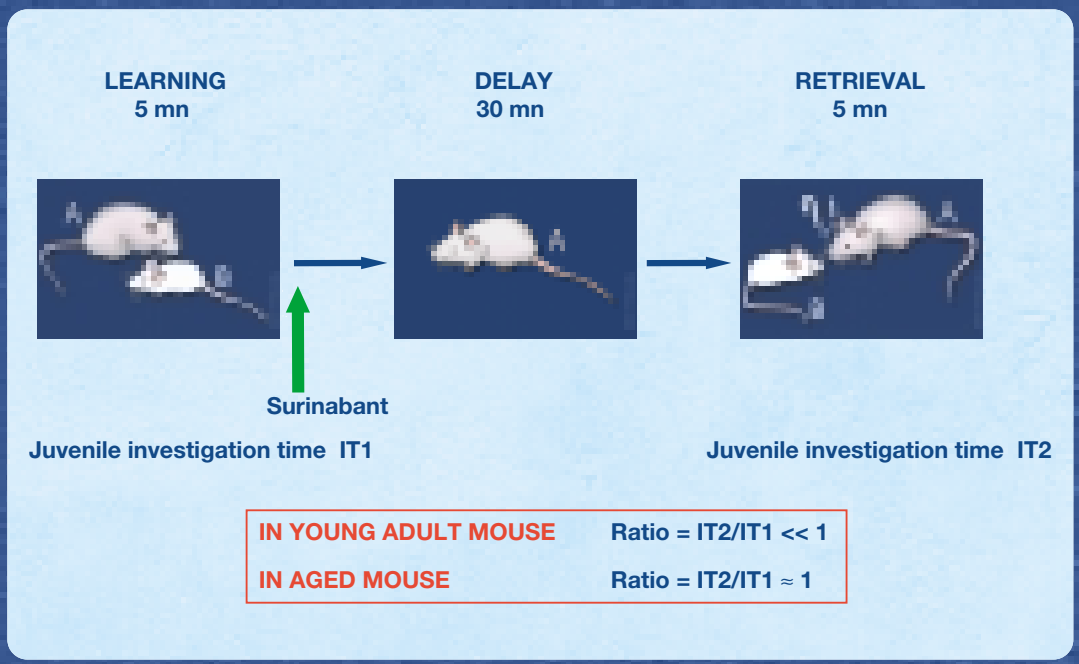
LEARNING 5 mn
DELAY 30 mn
RETRIEVAL 5 mn
Surinabant
Juvenile investigation time IT1
Juvenile investigation time IT2
IN YOUNG ADULT MOUSE Ratio = IT2/IT1 << 1
IN AGED MOUSE Ratio = IT2/IT1 ≈ 1

Figure 5 DOSE RELATIONSHIP OF SURINABANT

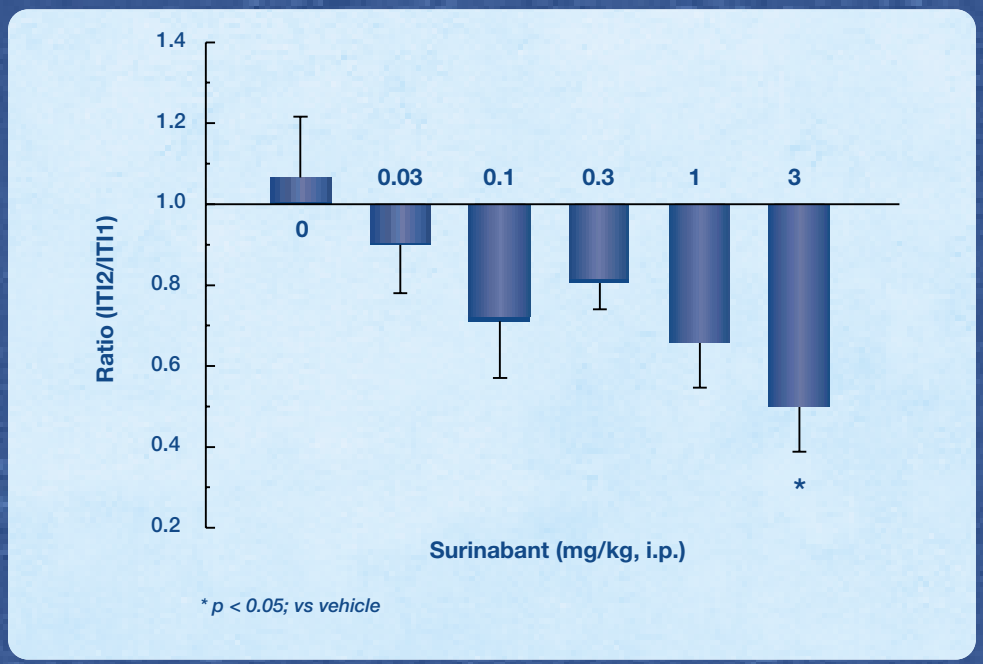
Ratio (IT2/IT1)
0 0.03 0.1 0.3 1 3
Surinabant (mg/kg, i.p.)
* p < 0.05; vs vehicle

Surinabant (3 mg/kg, i.p.) improves age-impaired spontaneous social recognition memory in mice

MORRIS WATER MAZE TASK IN RATS : EFFECT OF SURINABANT ON SPATIAL WORKING MEMORY IMPAIRED BY SCOPOLAMINE

Figure 6 EXPERIMENTAL DESIGN

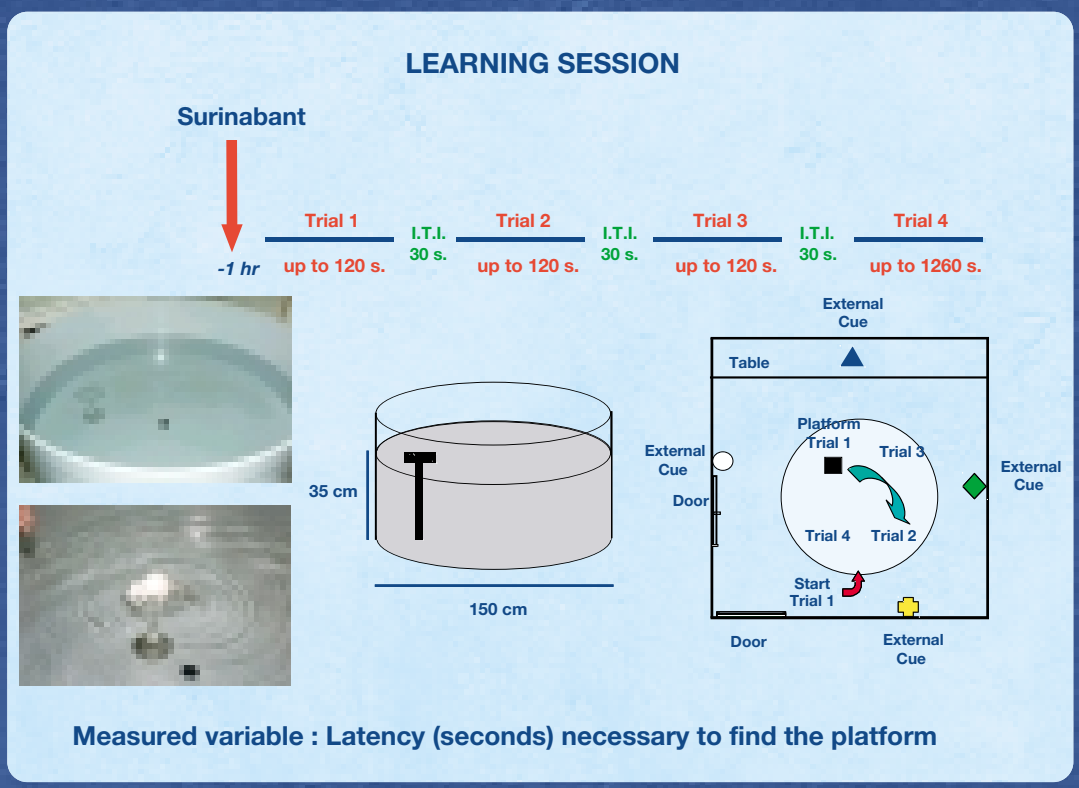
LEARNING SESSION
Surinabant
-1 hr
Trial 1 up to 120 s.
I.T.I. 30 s.
Trial 2 up to 120 s.
I.T.I. 30 s.
Trial 3 up to 120 s.
I.T.I. 30 s.
Trial 4 up to 1260 s.
35 cm
150 cm
Table
External Cue
Platform Trial 1
Trial 3
Trial 4
Trial 2
Start Trial 1
Door
Measured variable : Latency (seconds) necessary to find the platform

Figure 7 REVERSAL BY SURINABANT OF SCOPOLAMINE-INDUCED DEFICIT OF SPATIAL WORKING MEMORY

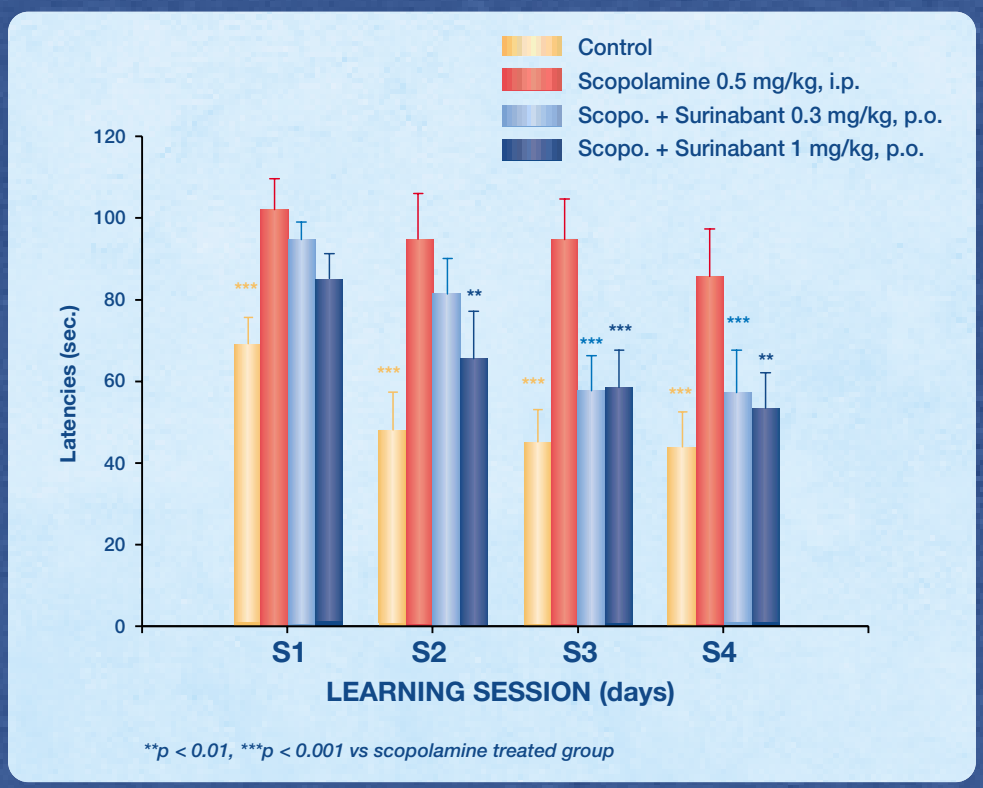
Control
Scopolamine 0.5 mg/kg, i.p.
Scopo. + Surinabant 0.3 mg/kg, p.o.
Scopo. + Surinabant 1 mg/kg, p.o.
Latencies (sec.)
S1 S2 S3 S4
LEARNING SESSION (days)
p < 0.01, *p < 0.001 vs scopolamine treated group

Surinabant (0.3-1 mg/kg, p.o.) fully restores a scopolamine-induced spatial working memory deficit in rats, an effect that becomes significant with repeated administrations

Conclusion

Surinabant a novel and selective CB1 receptor antagonist improves learning and memory functions in a variety of animal models :

- **Surinabant dose-dependently and fully reverses short-term episodic memory deficit induced by scopolamine in rats**
- **Surinabant displays a potent pro-cognitive activity in rats, improving long-term episodic memory**
- **Surinabant restores social memory recognition deficit induced by age in mice, an effect indicative of increased memory retention**
- **Surinabant reverses scopolamine-induced deficit of spatial working memory in rats**

Taken together, these results strongly suggest that Surinabant represents a promising drug candidate for the symptomatic treatment of cognitive disorders related to schizophrenia or Alzheimer's disease.

Acknowledgments

The expert technical assistance of D. LEBOUTEUX and C. PORSOLT was greatly appreciated

Presented at the XVth ICRS Symposium on Cannabinoids – Clearwater (Florida) – June 24-27, 2005